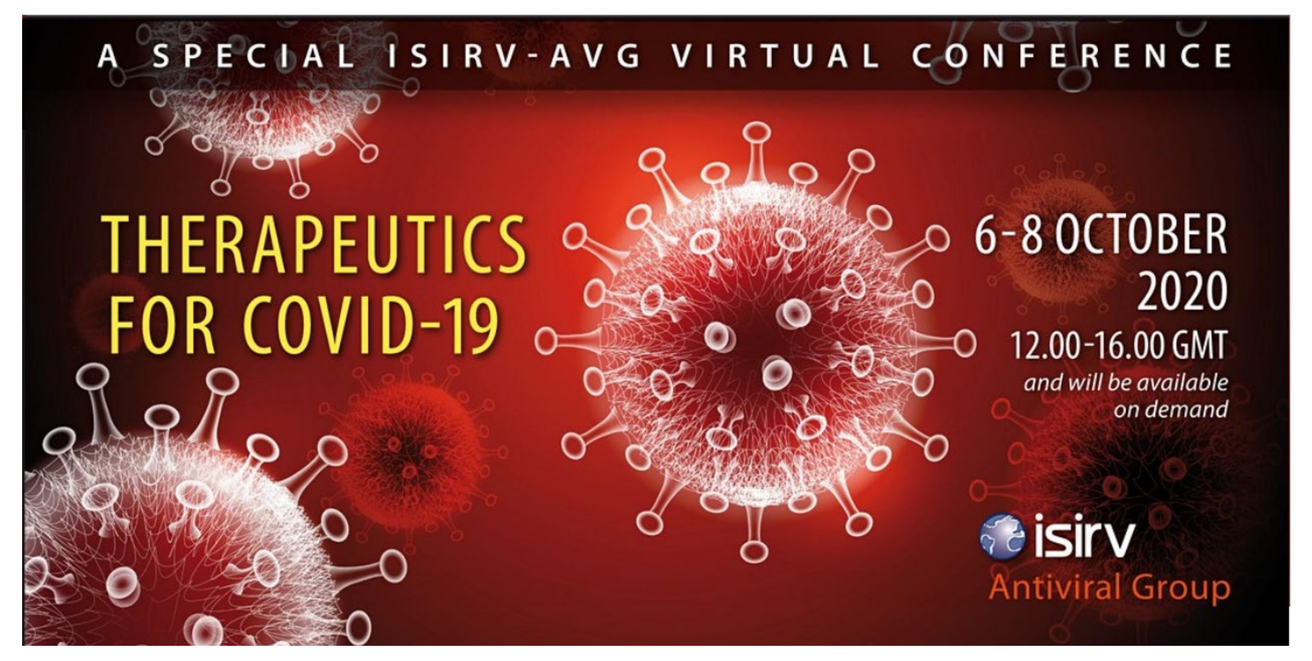
A SPECIAL ISIRV-AVG VIRTUAL CONFERENCE
THERAPEUTICS FOR COVID-19
6-8 OCTOBER 2020
12.00-16.00 GMT
and will be available on demand
isirv
Antiviral Group

Abstract # AAVGV0025

Evaluation of SARS-CoV-2 3C-like protease inhibitors using self-assembled monolayer desorption ionization mass spectrometry

Zachary A. Gurard-Levin[1], Cheng Liu[2], Andreas Jekle[2], Ruchika Jaisinghani[2], Suping Ren[2], Koen Vandyck[3], Dirk Jochmans[4], Pieter Leyssen[4], Johan Neyts[4], Lawrence M. Blatt[2], Leonid Beigelman[2], Julian A. Symons[2], Pierre Raboisson[3], Michael D. Scholle[1], and Jerome Deval[2]

[1]SAMDI Tech, Inc., Chicago, USA; [2]Aligos Therapeutics, Inc., South San Francisco, USA; [3]Aligos Belgium BV, Leuven, Belgium; [4]Rega Institute, KU Leuven, Leuven, Belgium ***Corresponding author: jdeval@aligos.com***

Background: **Severe acute respiratory syndrome coronavirus 2 (SARS-CoV-2) is the cause of the COVID-19 pandemic that began in 2019. The coronavirus 3-chymotrypsin-like cysteine protease (3CLpro) controls replication and is therefore considered a major target for antiviral discovery. Developing robust and sensitive in vitro screening assays is key to rapidly identify the most promising inhibitors of SARS-CoV-2 3CLpro.**

Methods: **SARS-CoV-2 3CLpro inhibitors were evaluated in a novel self-assembled monolayer desorption ionization mass spectrometry (SAMDI-MS) in parallel with a standard FRET enzymatic assay, with and without reducing agent. Antiviral potency of control compounds was also determined in cells infected with OC-43 or SARS-CoV-2. Potential for compound cytotoxicity was conducted in parallel with antiviral assessment (1).**

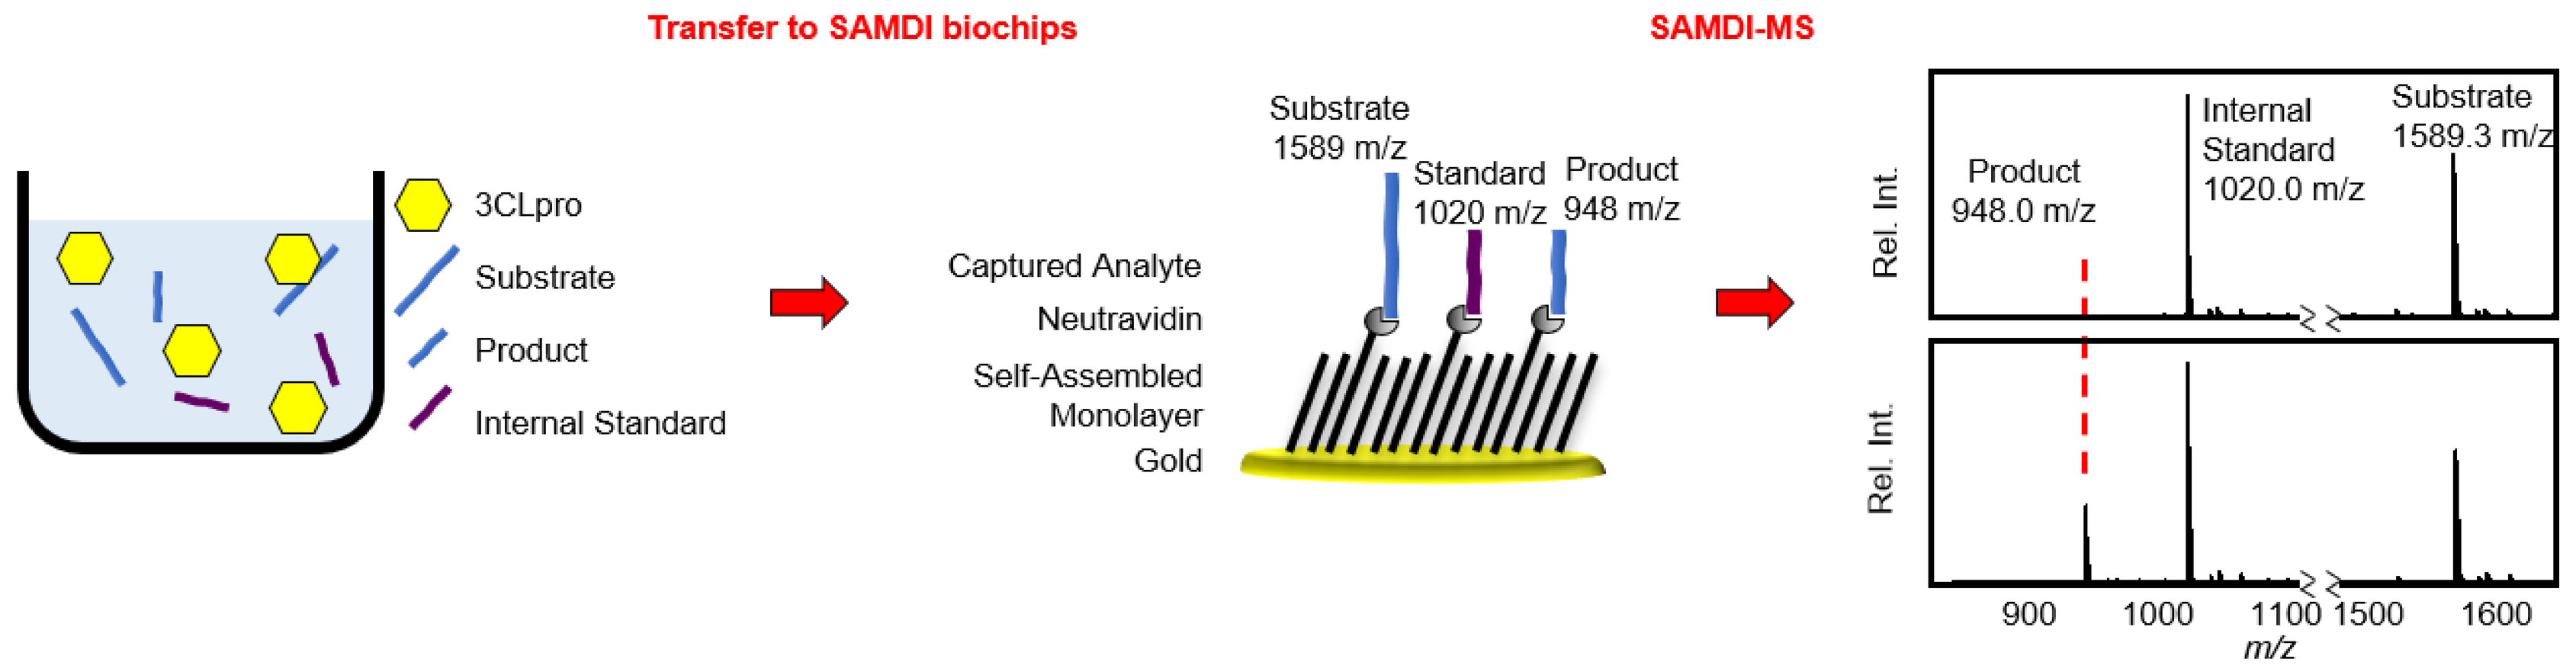
Transfer to SAMDI biochips
SAMDI-MS
3CLpro
Substrate
Product
Internal Standard
Captured Analyte
Neutravidin
Self-Assembled Monolayer
Gold
Substrate 1589 m/z
Standard 1020 m/z
Product 948 m/z
Product 948.0 m/z
Internal Standard 1020.0 m/z
Substrate 1589.3 m/z
Rel. Int.
900
1000
1100
1500
1600
m/z

Buffer optimization of SAMDI-MS assay

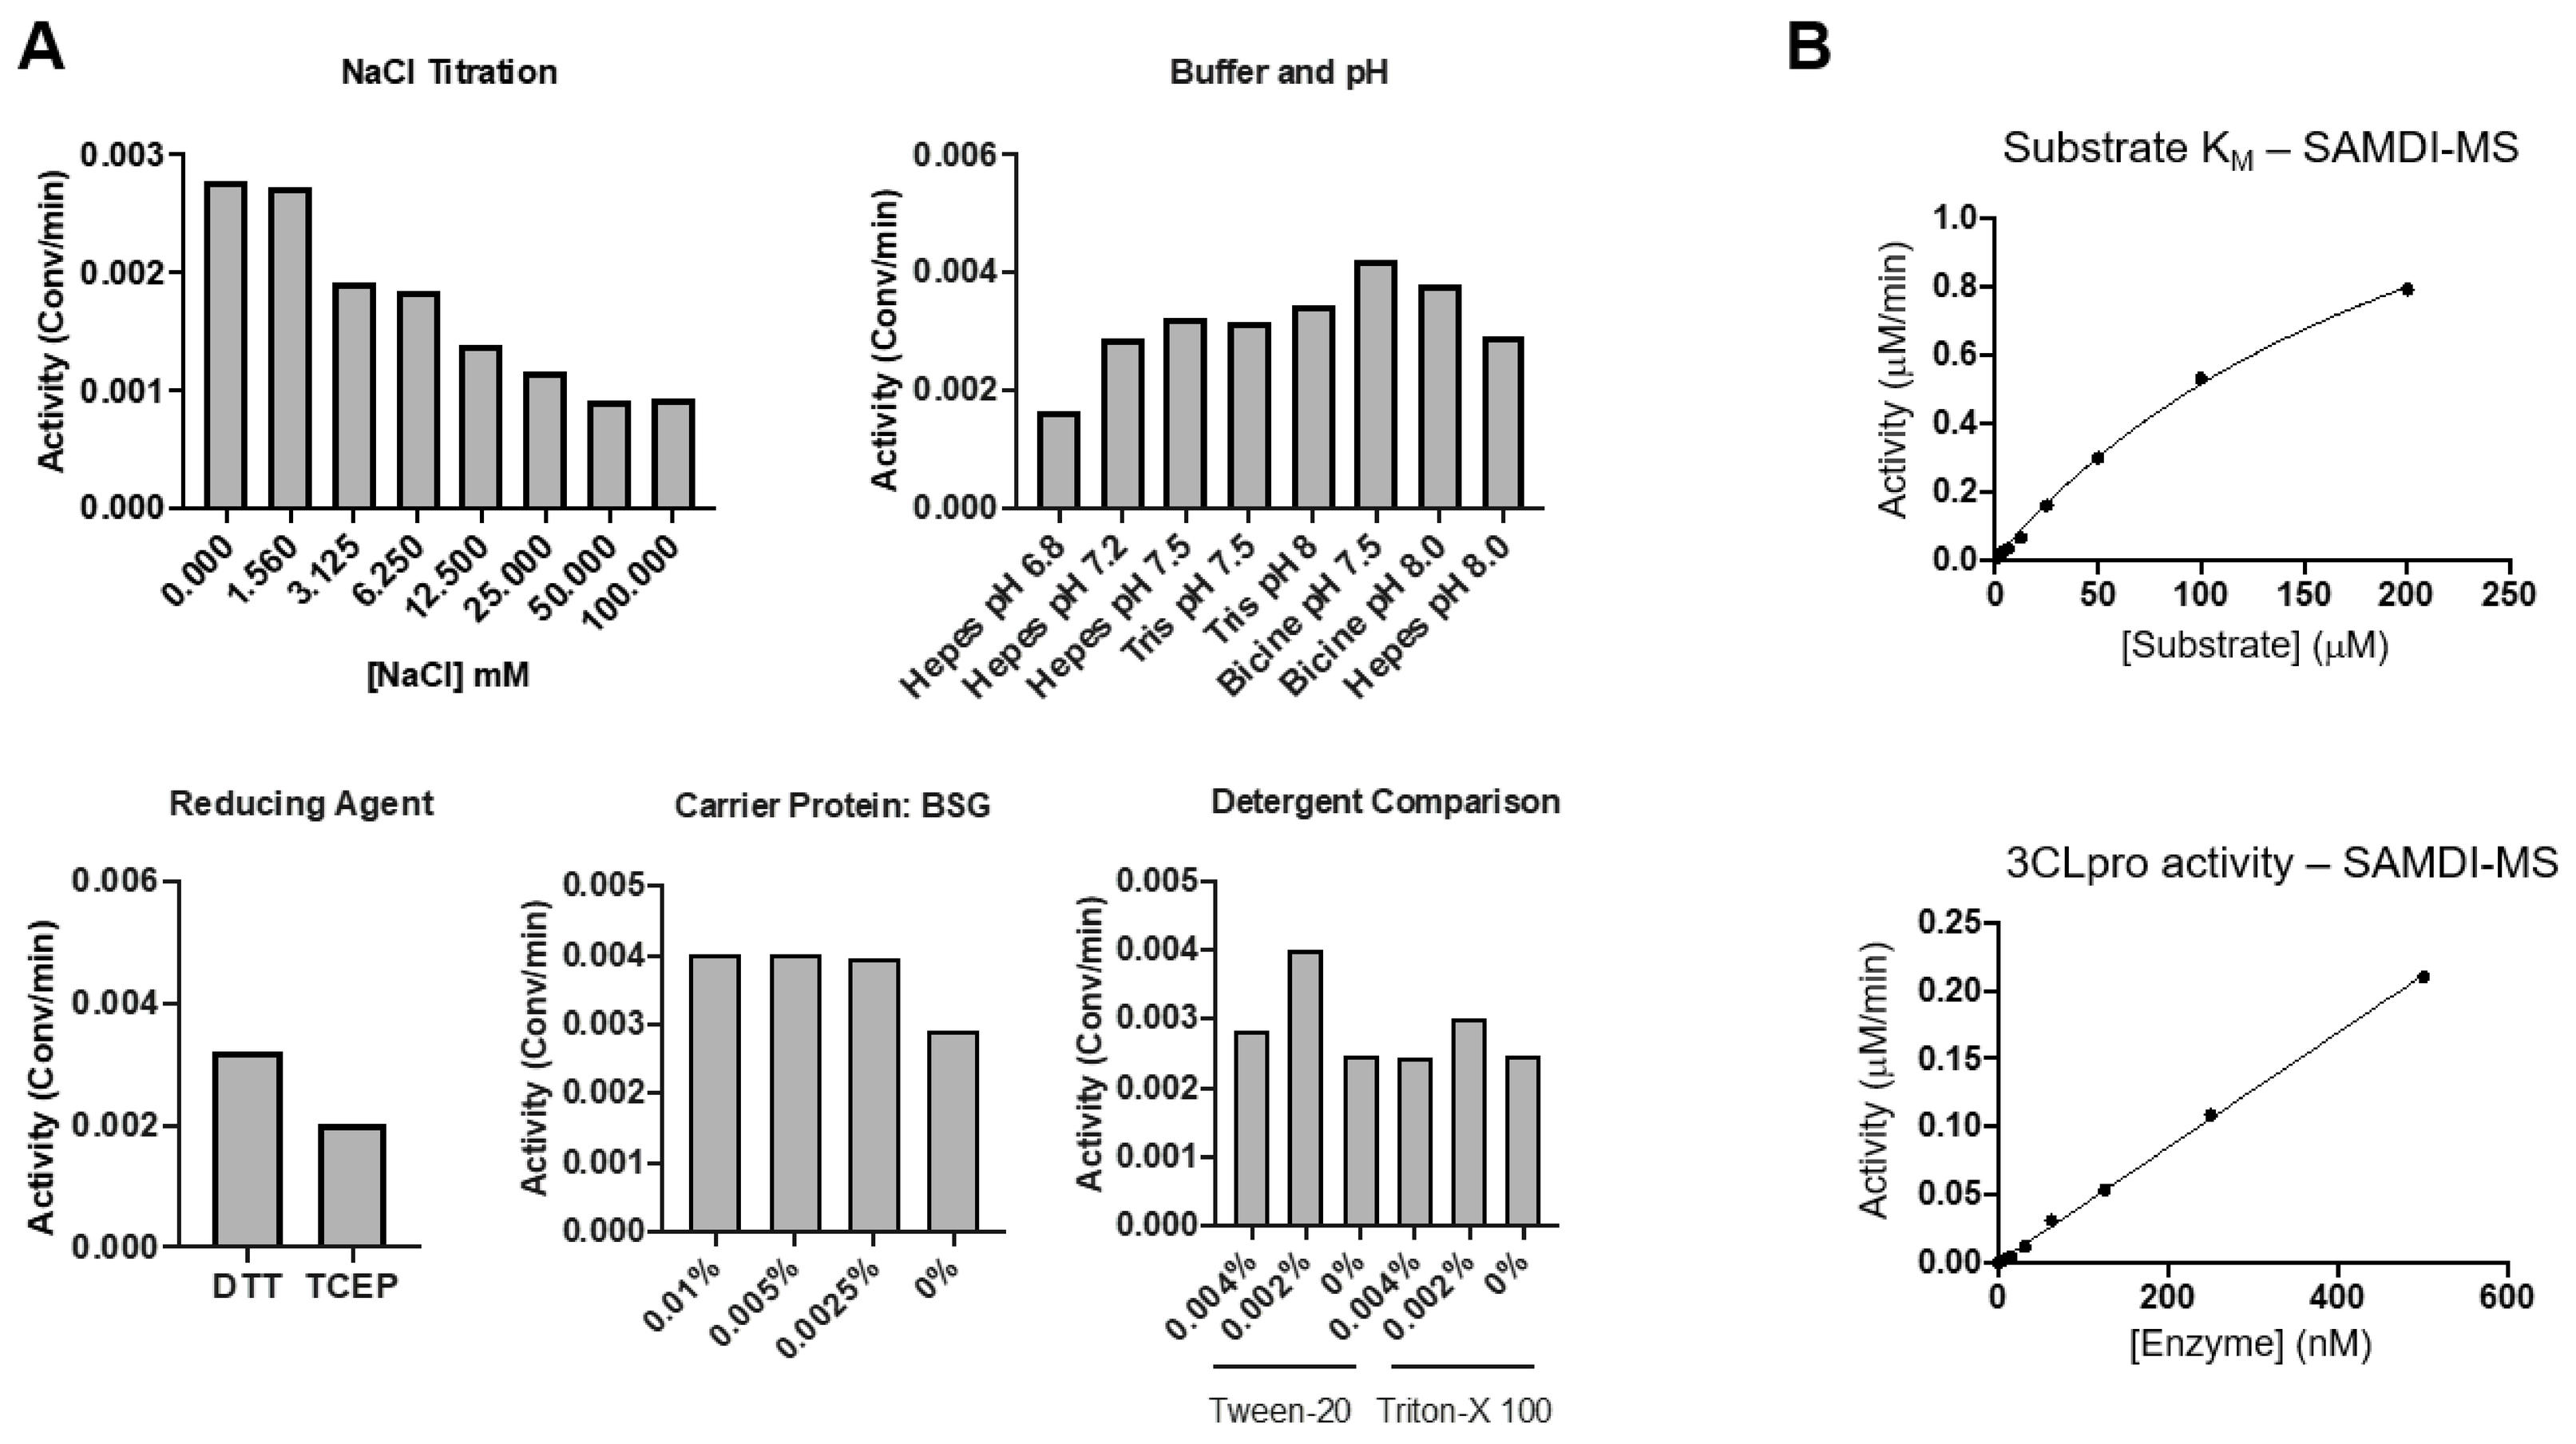
A
NaCl Titration
Activity (Conv/min)
[NaCl] mM
Buffer and pH
Hepes pH 6.8
Hepes pH 7.2
Hepes pH 7.5
Tris pH 7.5
Tris pH 8
Bicine pH 7.5
Bicine pH 8.0
Hepes pH 8.0
Reducing Agent
DTT
TCEP
Carrier Protein: BSG
Detergent Comparison
Tween-20
Triton-X 100
B
Substrate KM – SAMDI-MS
Activity (µM/min)
[Substrate] (µM)
3CLpro activity – SAMDI-MS
[Enzyme] (nM)

A) The initial velocity calculated from triplicate data utilizing the linear range of 3CLpro activity over a range of distinct conditions: NaCl concentration from 0 to 100 nM, buffer (Hepes, Tris, and Bicine) and pH (6.8–8.0), reducing agent (DTT and TCEP at 1 mM), bovine skin gelatin (BSG)—a carrier protein—concentration), and detergent Tween-20 and Triton-X 100. (B) K_M values were determined for the SAMDI Velocity of 3CLpro is linear over a range of concentrations in the SAMDI-MS assay using 10 µM substrate.

Robustness of the SAMDI-MS assay

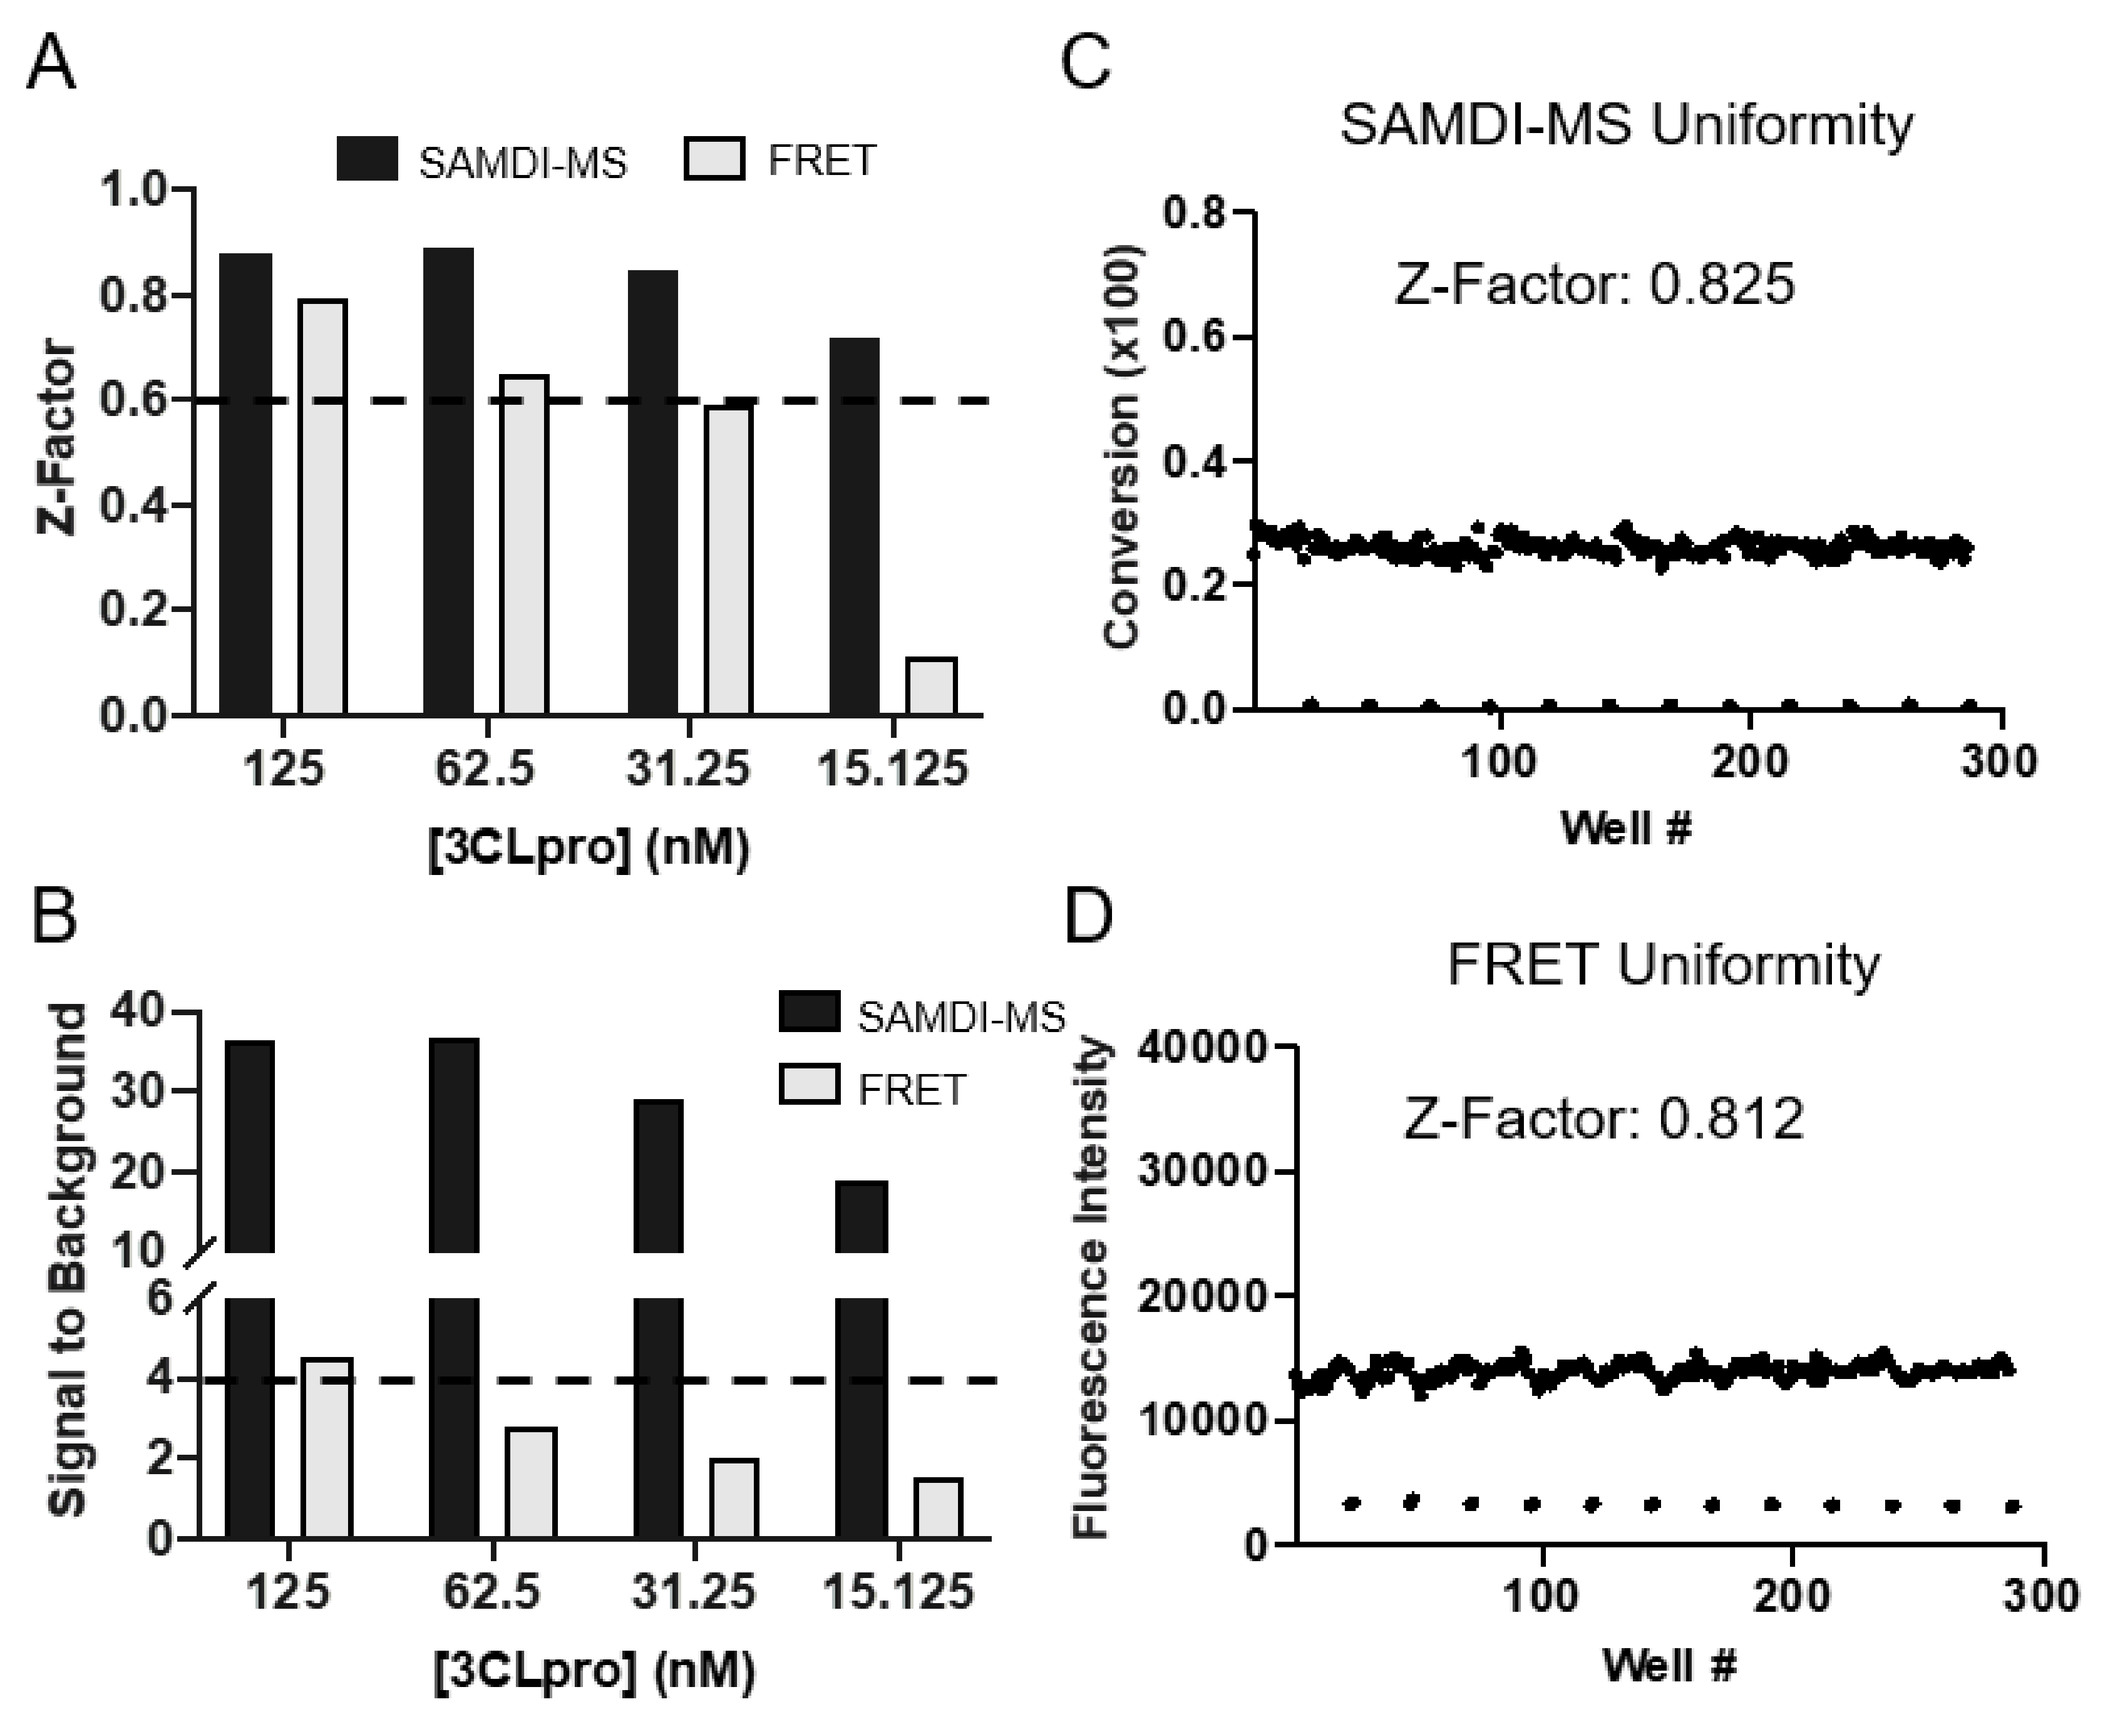
A
SAMDI-MS
FRET
Z-Factor
[3CLpro] (nM)
B
Signal to Background
C
SAMDI-MS Uniformity
Z-Factor: 0.825
Conversion (x100)
Well #
D
FRET Uniformity
Z-Factor: 0.812
Fluorescence Intensity

- **(A) Z-factor and signal to background ratios (B) for SAMDI-MS and FRET assays under different enzyme concentrations. The dotted line indicates an assay robustness cutoff of 0.6 for Z-factor, and 4-fold for signal to background. Full plate uniformity data using optimized conditions and 6 µL reaction volumes for SAMDI-MS (C) and FRET (D) assays.**

Comparison of 3CLpro inhibition between FRET and SAMDI-MS

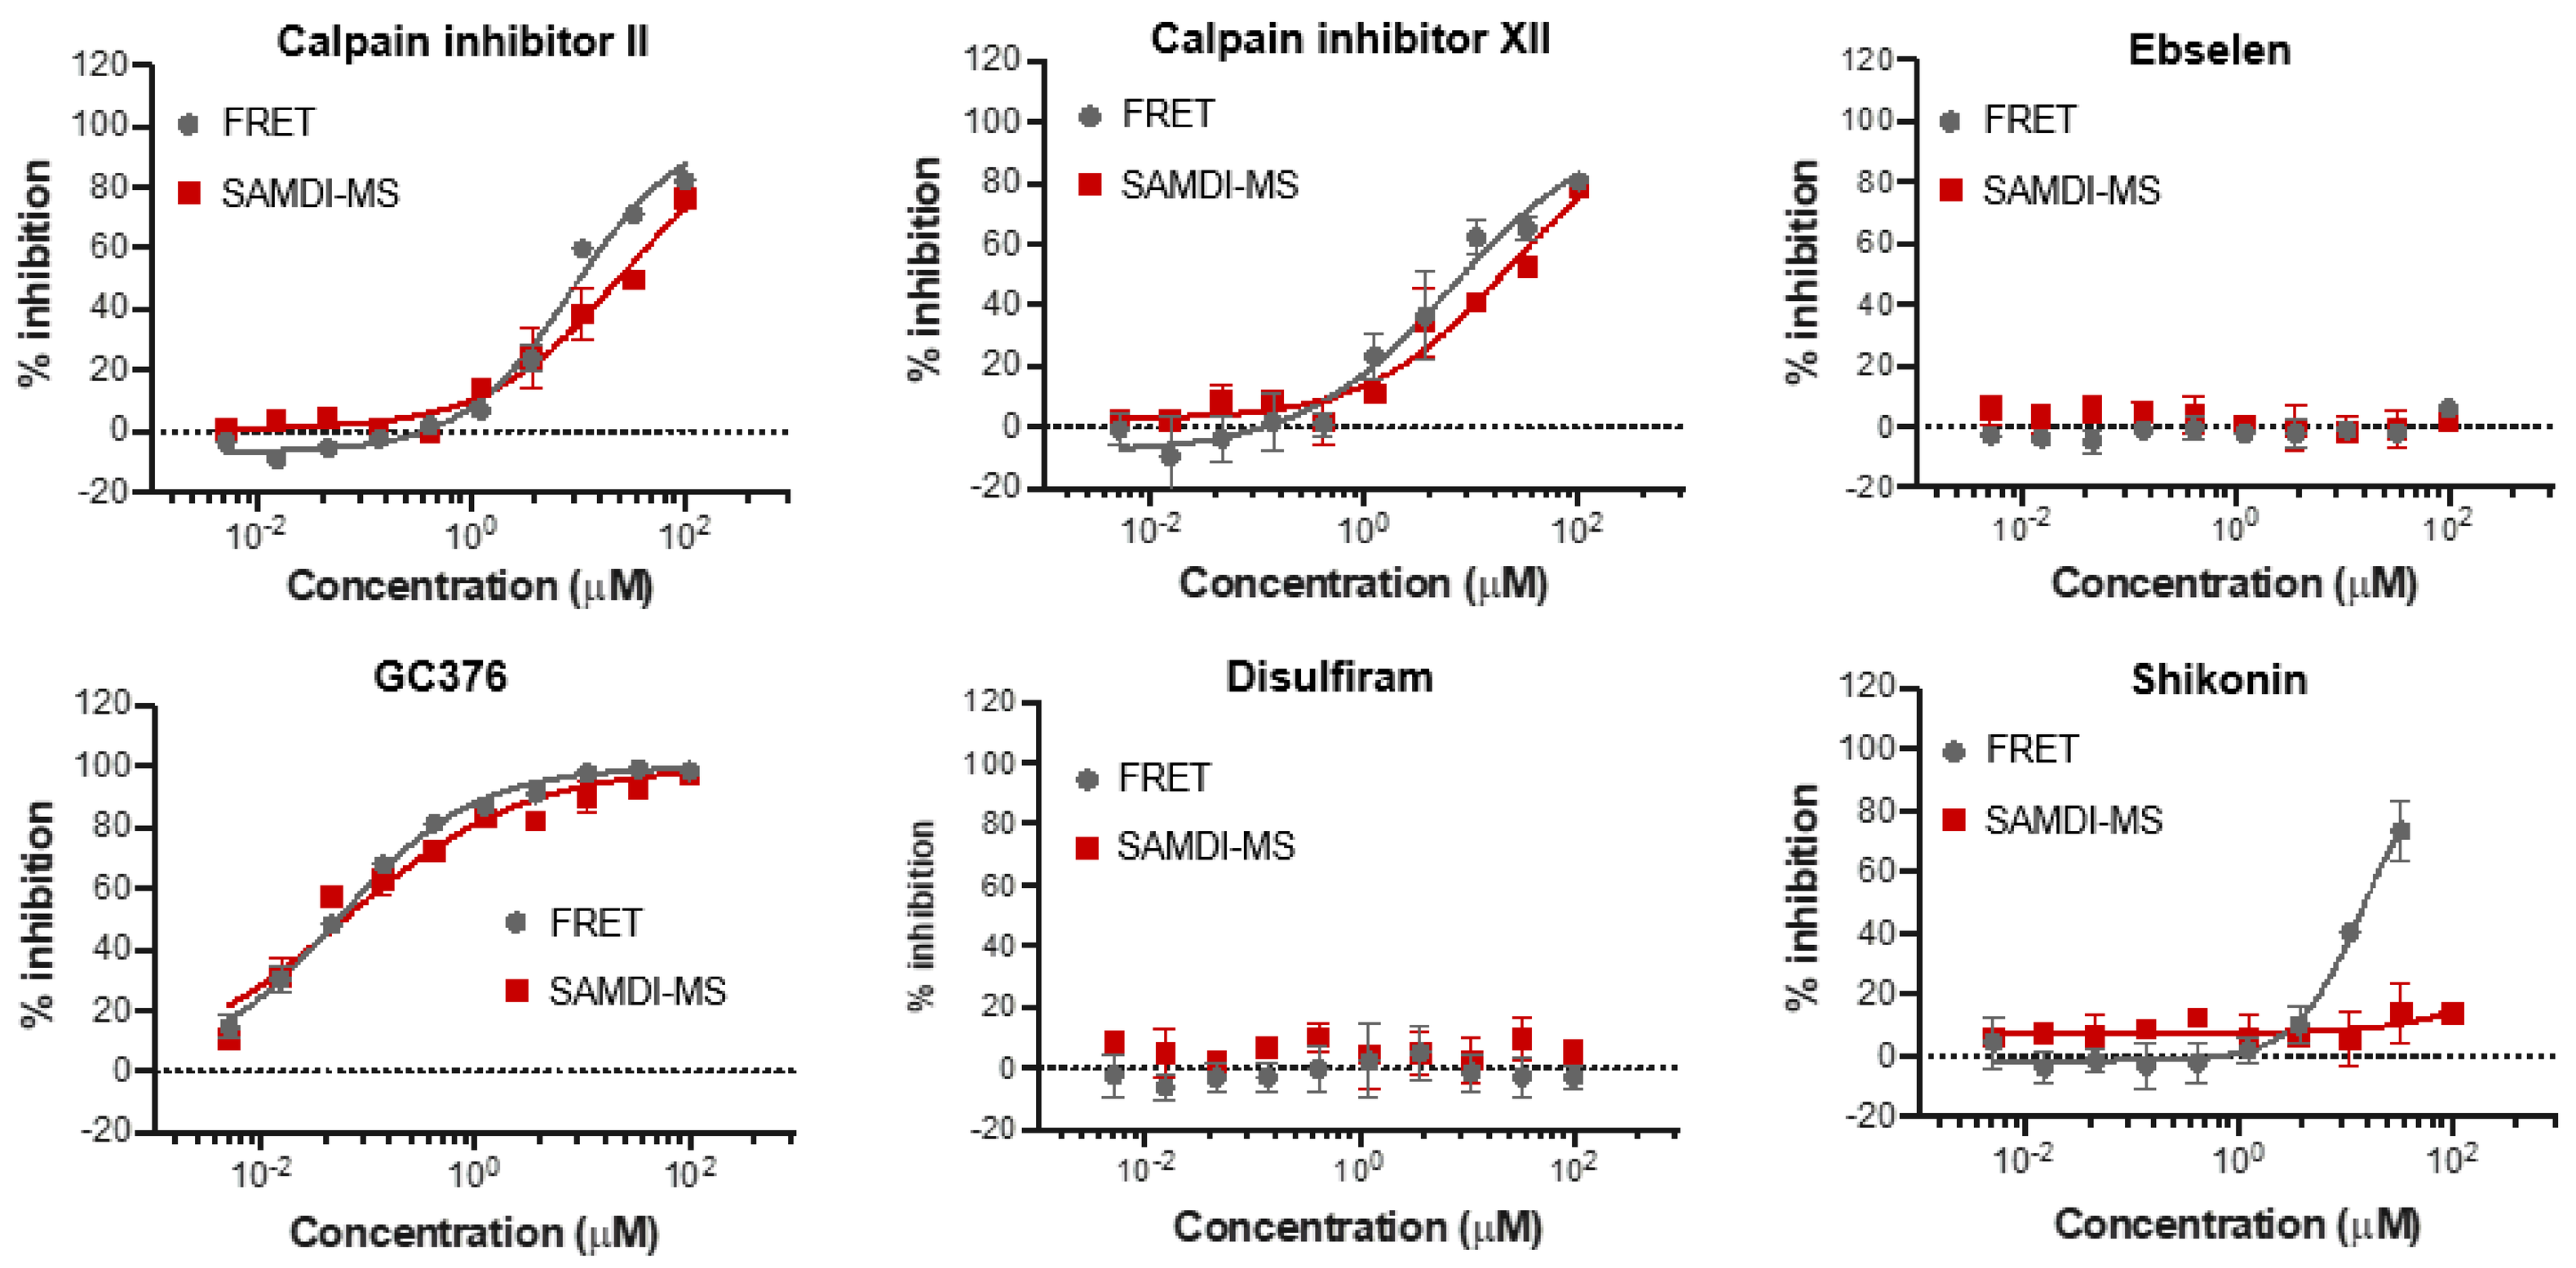
Calpain inhibitor II
Calpain inhibitor XII
Ebselen
GC376
Disulfiram
Shikonin
FRET
SAMDI-MS
% inhibition
Concentration (µM)

IC_{50} measurements of six reported 3CLpro inhibitors were calculated in the SAMDI-MS (red) and FRET (grey) assay formats. Experiments were performed in duplicate and error bars represent standard deviation.

Effect of reducing conditions on inhibition potency

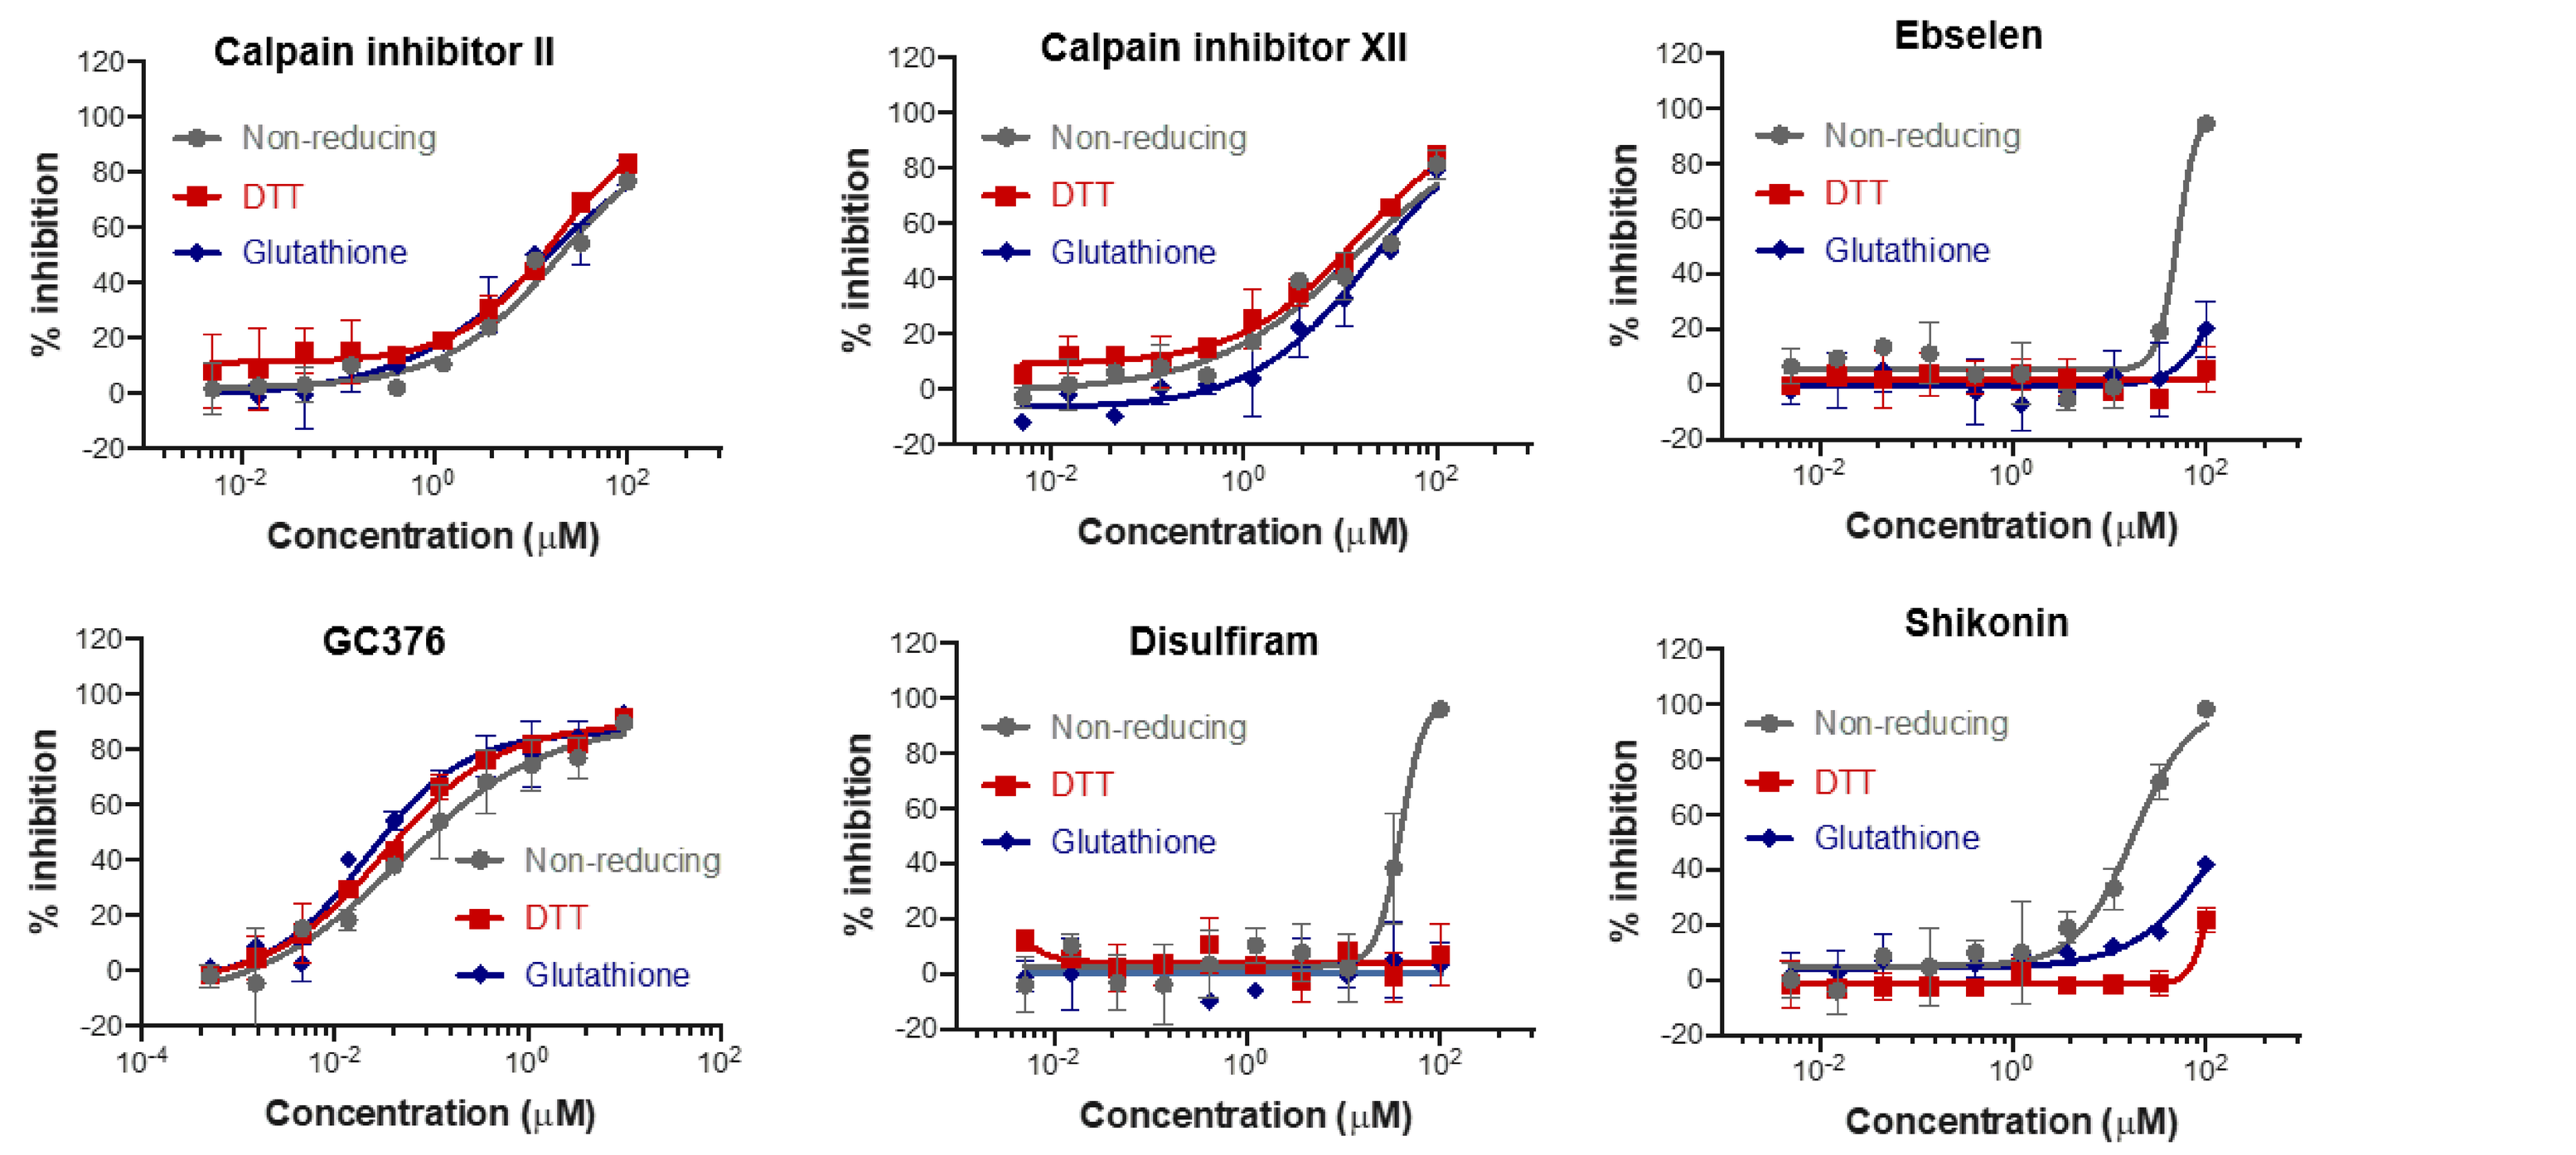
Calpain inhibitor II
Calpain inhibitor XII
Ebselen
GC376
Disulfiram
Shikonin
Non-reducing
DTT
Glutathione
% inhibition
Concentration (µM)

IC_{50} measurements of 6 reported 3CLpro inhibitors measured by SAMDI-MS in the absence of reducing agent (grey), in the presence of 1 mM DTT (red), and in the presence of 1 mM glutathione (blue). Experiments were performed in duplicate and error bars represent standard deviation.

Summary of antiviral activity against OC43-CoV and SARS-CoV-2

Test Article	OC-43 Hela Cells		OC-43 MRC-5 Cells		SARS-CoV-2 VeroE6 Cells	
	EC_{50} (µM)	CC_{50} (µM)	EC_{50} (µM)	CC_{50} (µM)	EC_{50} (µM)	CC_{50} (µM)
Remdesivir	0.114±0.009	>1	0.21	≥50	n.d.	n.d.
GS-441524	n.d.	n.d.	n.d.	n.d.	<0.8	55±18
GC376	0.42±0.033	>10	0.83	>50	10±4.2	>100
Calpain Inhibitor II	20.7±3.3	>100	20.95	>100	27±1.4	>100
Calpain Inhibitor XII	15.2±2.4	60.3±8.3	6.93	>50	1.3±0.57	27±0.0
Ebselen	>100	15.9±2.4	>100	8.23	>100	37.5±9.2
Disulfiram	>100	75	>100	8.35	>100	13.5±0.70
Shikonin	>100	1.4±0.001	>100	0.32	>100	1.55±0.07

Results and Conclusions

The label-free SAMDI-MS assay was optimized and validated with known inhibitors of coronavirus 3CLpro such as GC376 (IC_{50}= 0.060 µM), calpain inhibitors II and XII (IC_{50} ~20-25 µM). The FDA-approved drugs shikonin, disulfiram, and ebselen did not inhibit SARS-CoV-2 3CLpro activity in the SAMDI-MS assay under physiologically relevant reducing conditions. The three drugs did not directly inhibit human β-coronavirus OC-43 or SARS-CoV-2 in vitro, but instead induced cell death. Compared with a traditional FRET readout, the SAMDI-MS assay offers greater sensitivity and eliminates false positive inhibition from compound interference with the optical signal. In conclusion, the SAMDI-MS 3CLpro assay, combined with antiviral and cytotoxic assessment, provides a robust platform to evaluate antiviral agents directed against SARS-CoV-2.

References: Gurard-Levin ZA, Liu C, Jekle A, Jaisinghani R, Ren S, Vandyck K, Jochmans D, Leyssen P, Neyts J, Blatt LM, Beigelman L, Symons JA, Raboisson P, Scholle MD, Deval J. Antiviral Res. 2020 Sep 5;182:104924. doi: 10.1016/j.antiviral.2020.104924.